Evaluation of the Efficacy of PeriphaGen Neurotrophin-3 Expressing Vector for Oxaliplatin Induced Peripheral Neuropathy

Fumagalli G[a], Chiorazzi A[a], Meregalli C[a], Carozzi VA[a], Monza L[c], Pozzi E[a], Alberti P[a,b], Goss J[d], Krisky D[d], Wechuck J[d], Marmiroli P[a], Cavaletti G[a].

a. University of Milano-Bicocca, Experimental Neurology Unit, School of Medicine and Surgery, Monza, Italy
b. PhD Program in Neuroscience, University of Milano-Bicocca
c. PhD Program in Translational and Molecular Medicine, DIMET, University of Milano-Biocca
d. PeriphaGen Inc. Pittsburgh, PA 15203

g.fumagalli14@campus.unimib.it

Background and Aim of the study

Oxaliplatin (OHP) is one of the most widely used anticancer drugs for the treatment of colorectal cancer. Despite its efficacy, its clinical use is often limited by the onset of Peripheral Neurotoxicity (OIPN) and neuropathic pain.
Neurotrophin 3 (NT3) is able to modulate the onset of neuropathic pain with effects on proprioceptive and nociceptive neurons and to promote growth and neuronal survival; it also plays an important role in remyelination and nerve regeneration.
The purpose of this study was to verify if PGN-503 (NT3-expressing herpes simplex virus based vector) is able to reduce the severity of OIPN.

Experimental design

40 female Balb/c mice were randomized into 4 experimental groups of 10 animals each: untreated animals (CTRL); animals treated with OHP; animals treated with OHP and injected with placebo vector; animals treated with OHP and injected with PGN-503 vector. OHP was administered intravenously 3.5mg/kg, twice/week for 4 weeks; placebo and PGN-503 vector (25µL) were administered subcutaneously into the plantar surface of the hind paws 3 days before the first OHP injection. The groups were observed at 3, 6 and 9 weeks of follow–up (FU).

Results

Neurophysiological examination

Neurophysiological recordings were performed in digital nerves at baseline, at end of treatment, after 3, 6 and 9 weeks of follow up

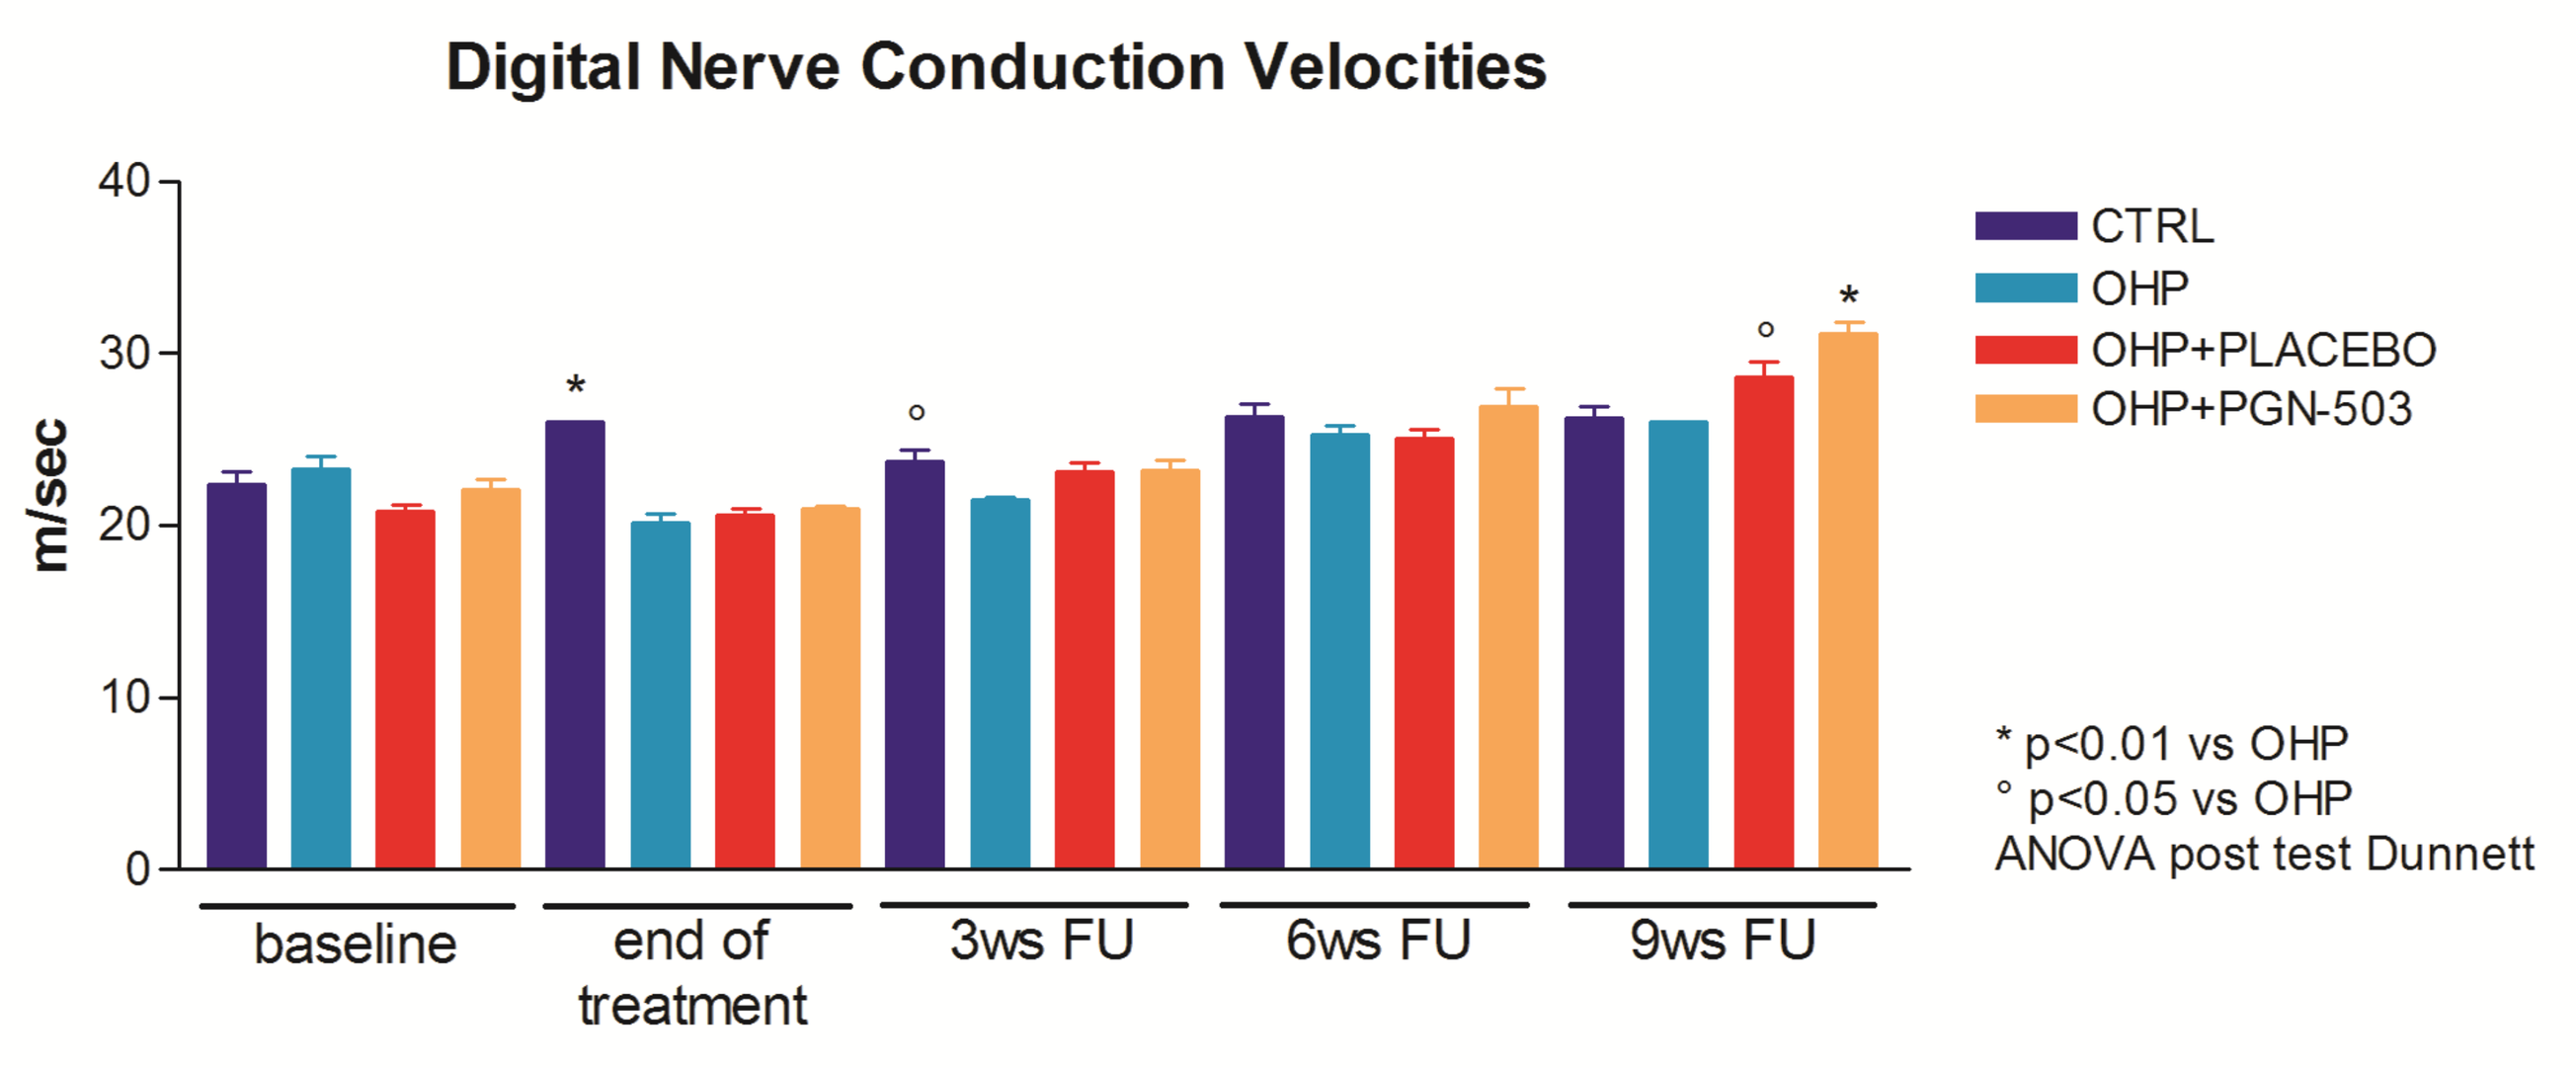
Digital Nerve Conduction Velocities
m/sec
CTRL
OHP
OHP+PLACEBO
OHP+PGN-503
* p<0.01 vs OHP
° p<0.05 vs OHP
ANOVA post test Dunnett
baseline
end of treatment
3ws FU
6ws FU
9ws FU

Digital Nerve Conduction Velocities (NCV)

At the end of treatment OHP induced a significant reduction in digital conduction velocity compared to control mice. PGN-503 mice demonstrated a significant recovery in digital conduction velocity by 6 weeks FU while OHP or placebo groups recovered by 9 weeks FU.

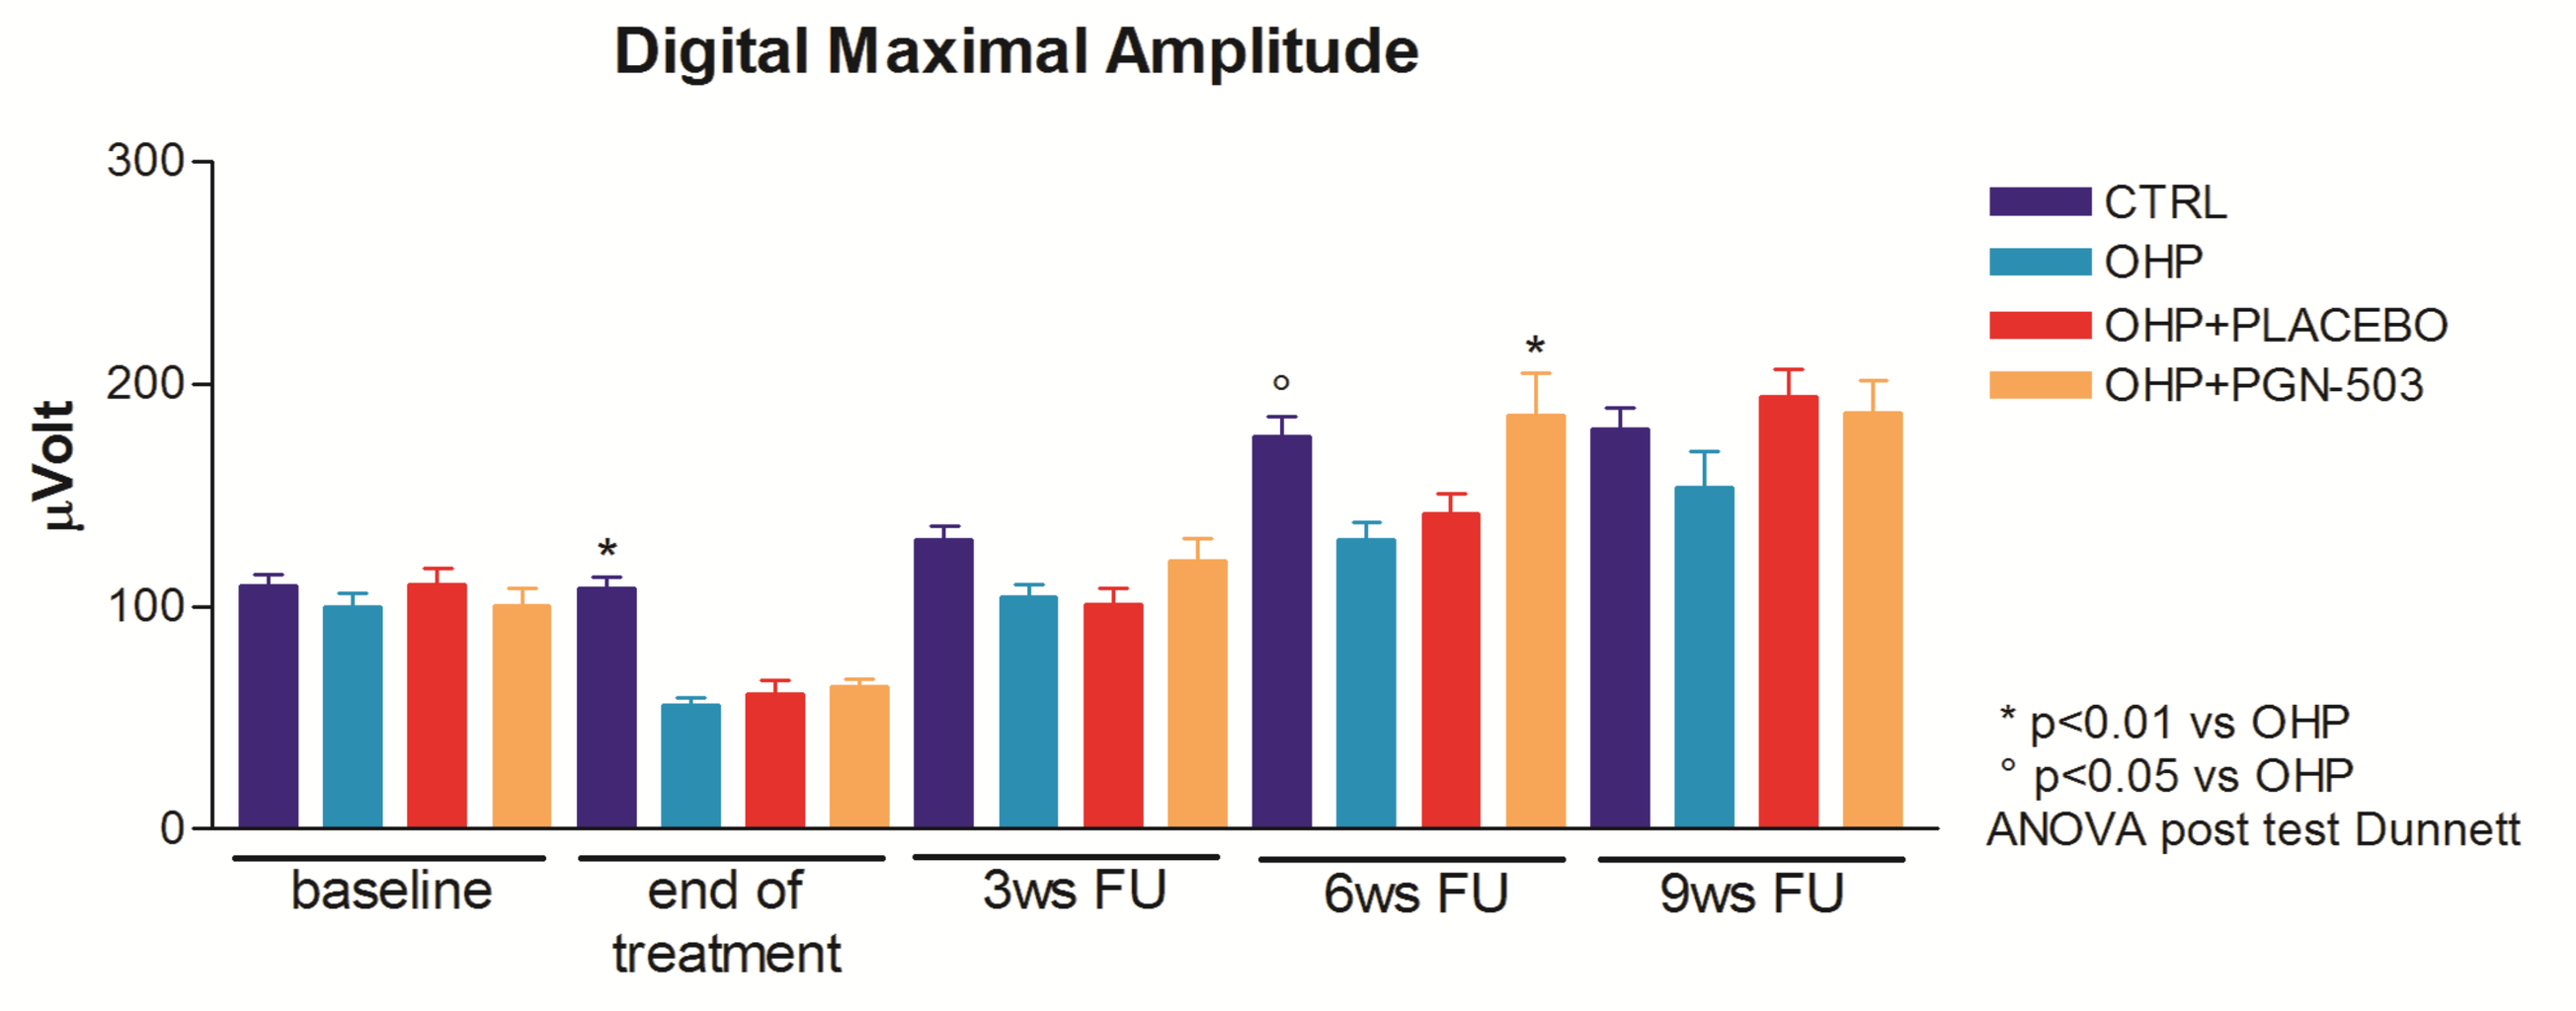
Digital Maximal Amplitude
µVolt
CTRL
OHP
OHP+PLACEBO
OHP+PGN-503
* p<0.01 vs OHP
° p<0.05 vs OHP
ANOVA post test Dunnett
baseline
end of treatment
3ws FU
6ws FU
9ws FU

Digital maximal amplitude (AMP)

At the end of treatment OHP induced a significant reduction in digital amplitude in all groups compared to control mice.
PGN-503 mice showed a significant recovery in digital amplitude by 3 weeks FU not observed in the OHP or placebo groups, which recovered by 9 weeks FU.

Behavioural tests

Animals were tested for sensory threshold to the mechanical (Dynamic Test) and thermal (Plantar Test) stimulation at baseline, at end of treatment, after 3, 6 and 9 weeks of follow up

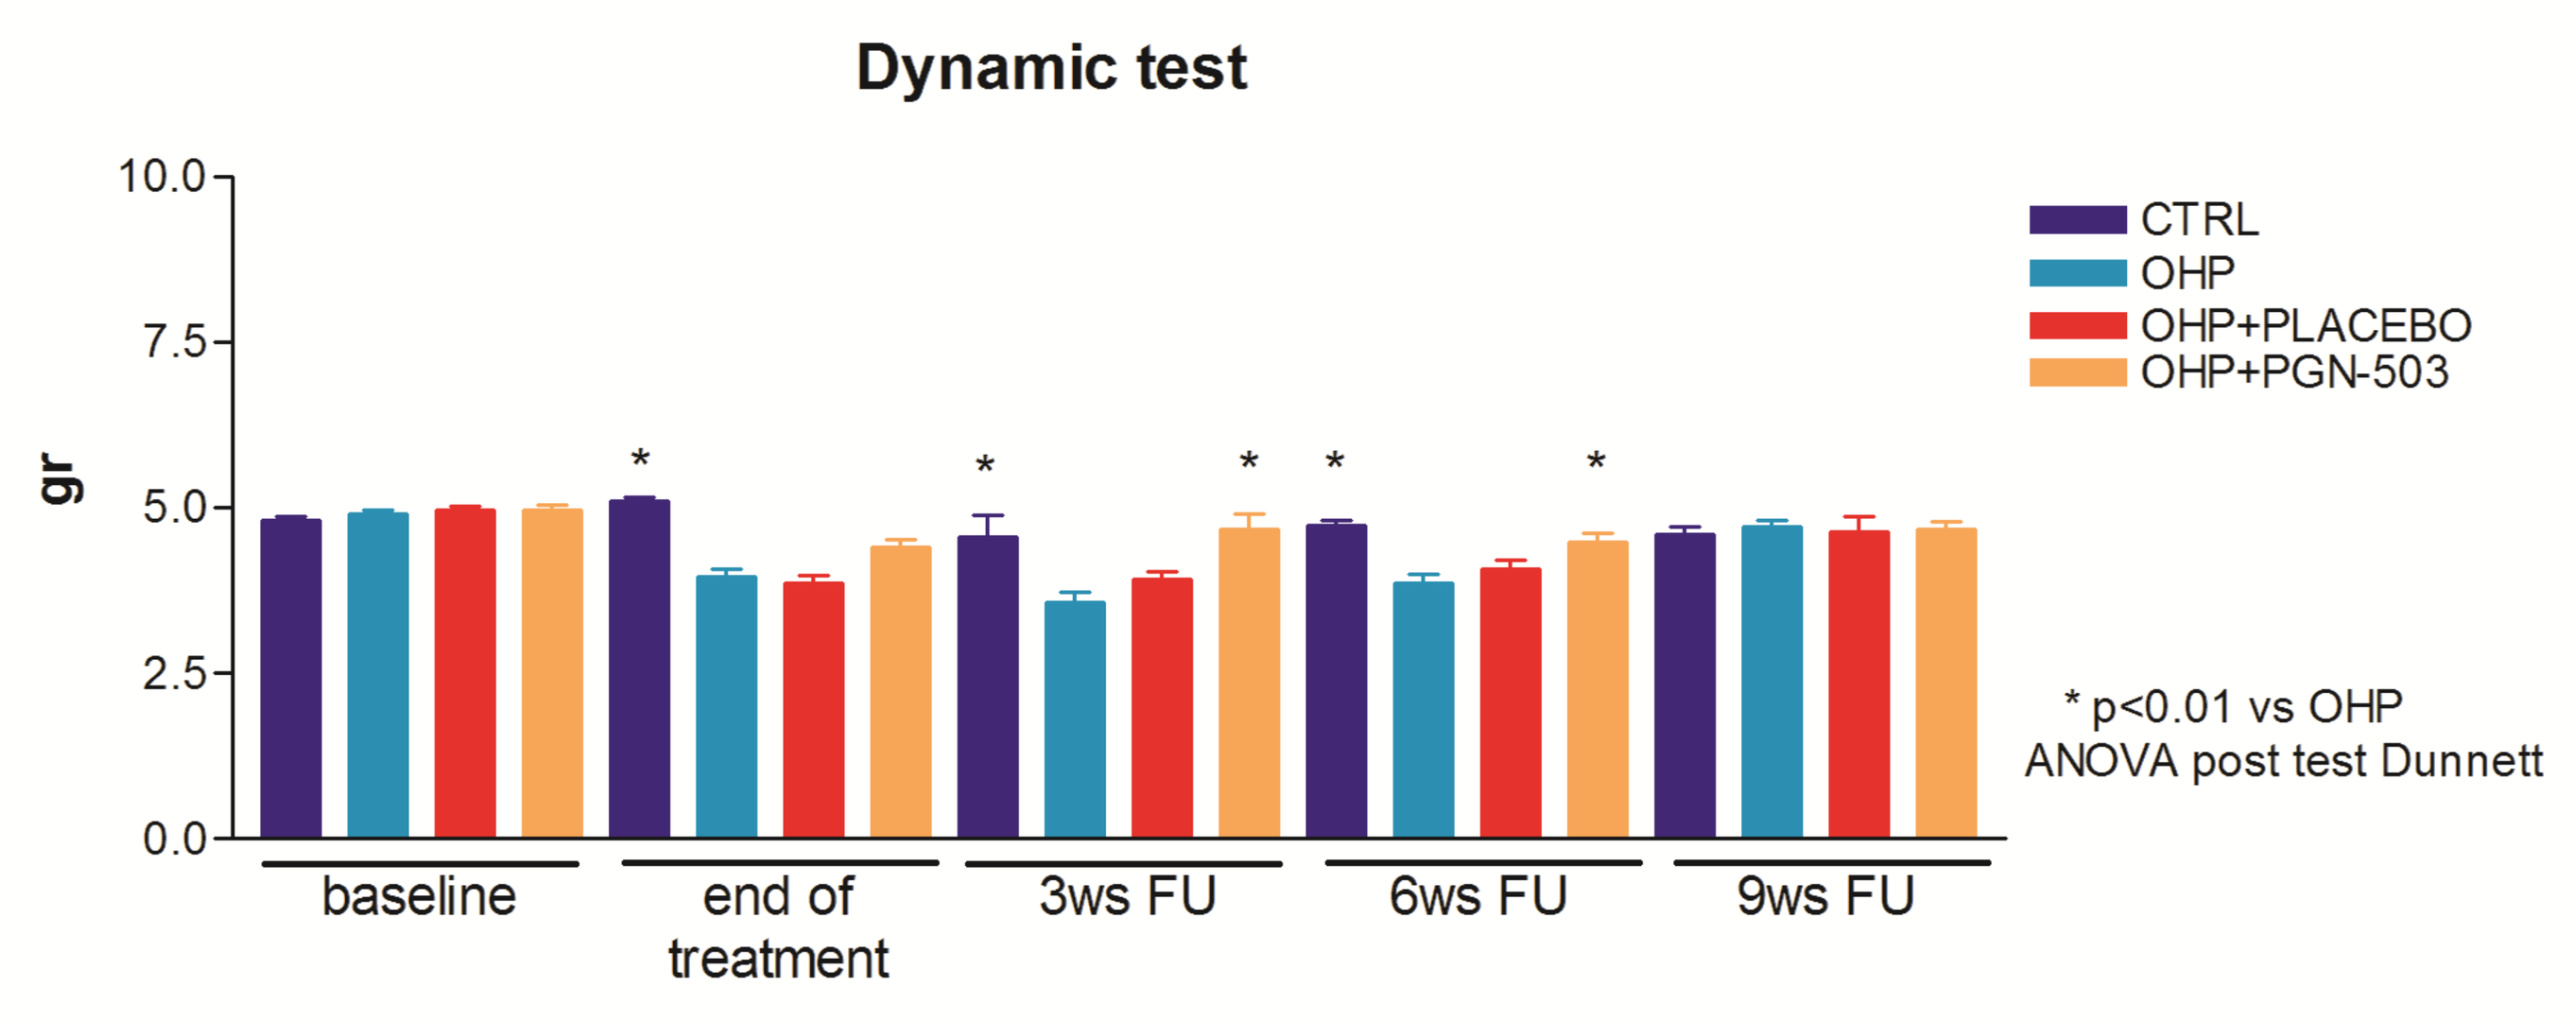
Dynamic test
gr
CTRL
OHP
OHP+PLACEBO
OHP+PGN-503
* p<0.01 vs OHP
ANOVA post test Dunnett
baseline
end of treatment
3ws FU
6ws FU
9ws FU

Dynamic test

Mechanical allodynia was observed in all OHP groups at the end of treatment, but it was less severe in the PGN-503 mice. Mechanical allodynia was completely recovered in the PGN-503 mice at week 3 FU whereas at week 9 FU in the OHP and placebo groups.

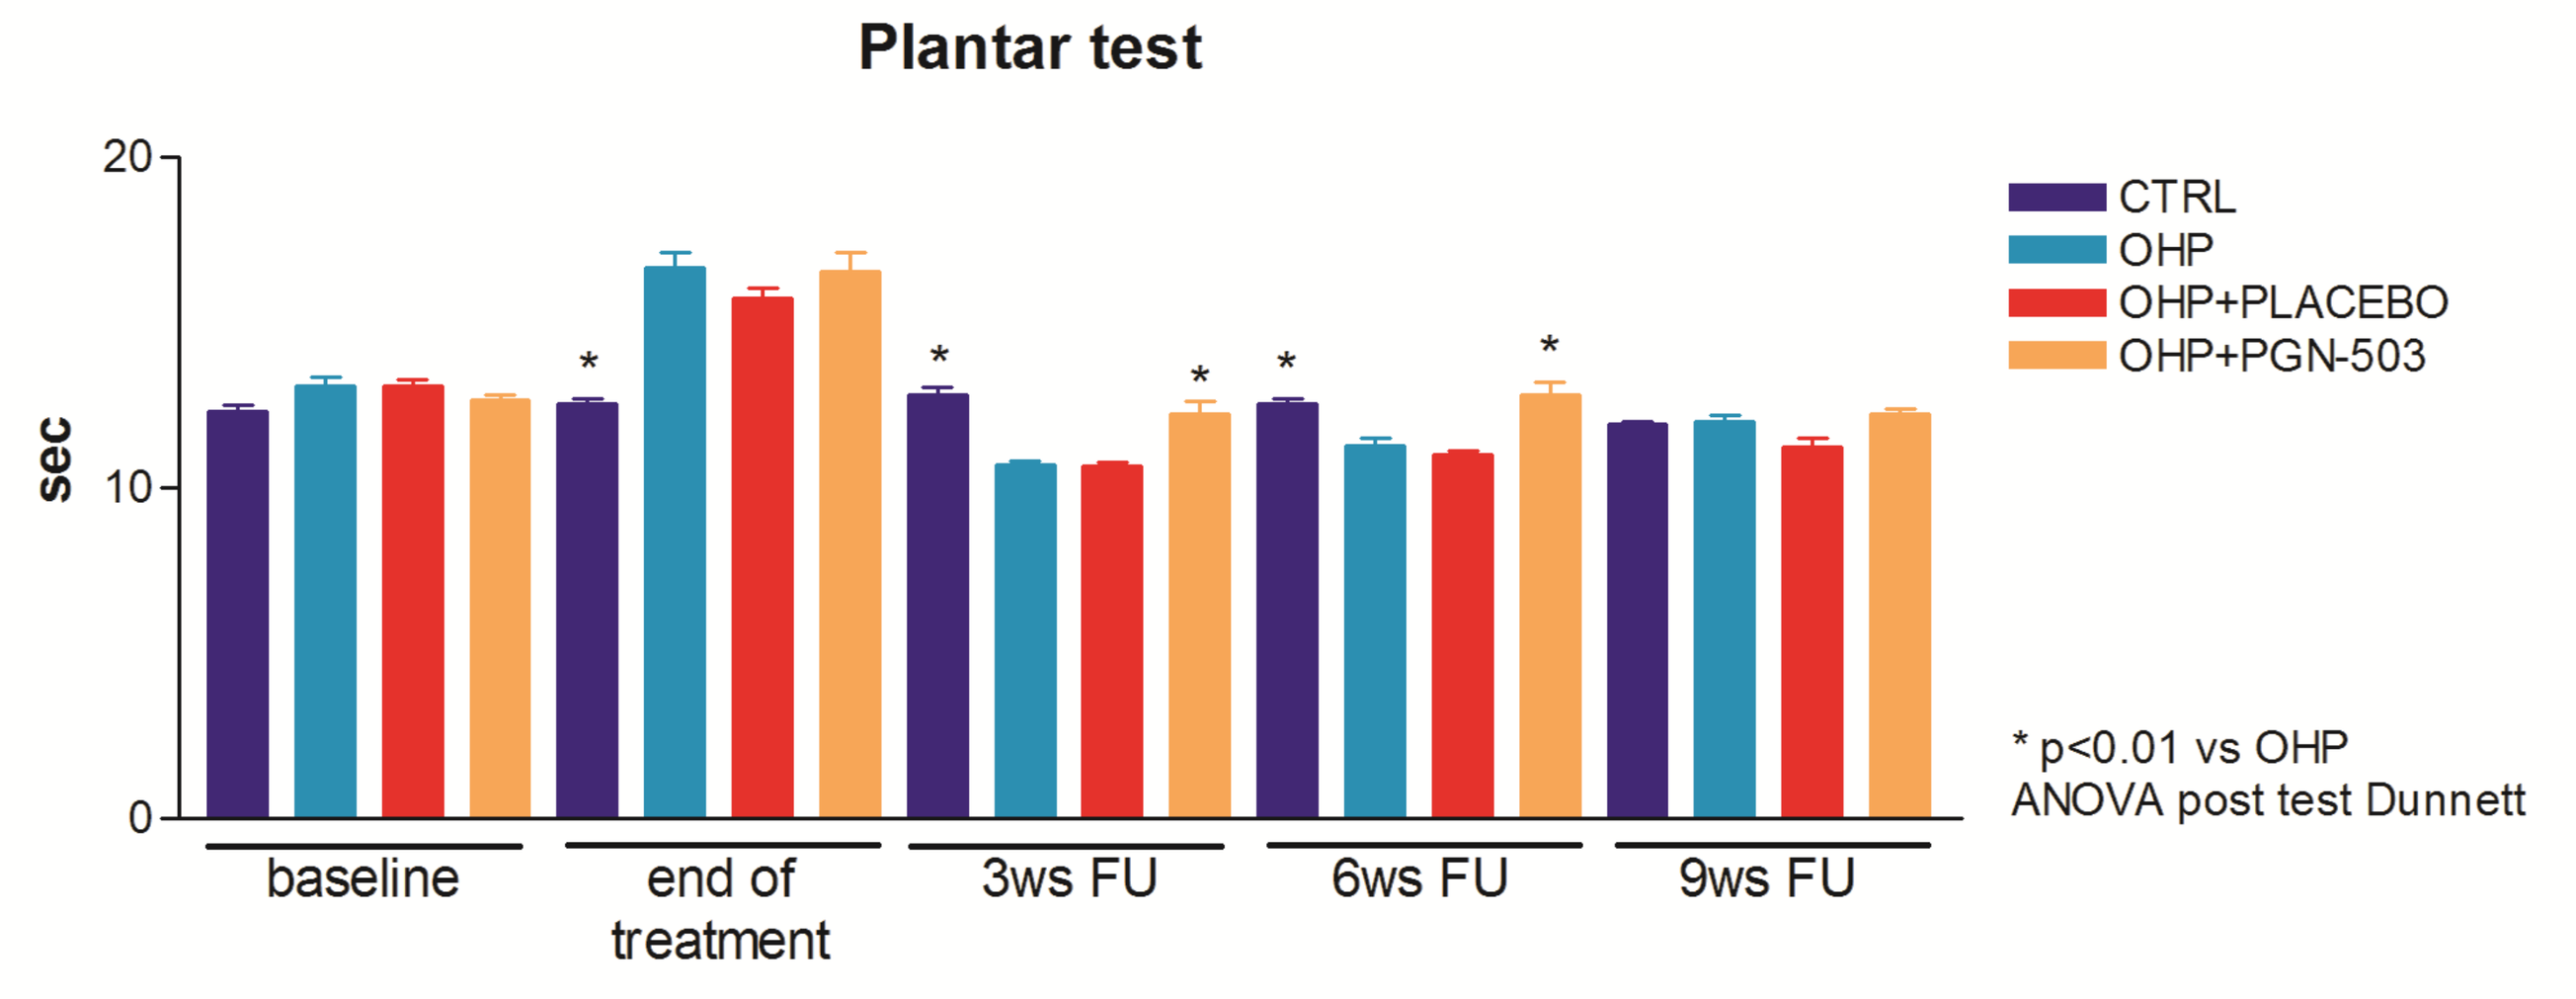
Plantar test
sec
CTRL
OHP
OHP+PLACEBO
OHP+PGN-503
* p<0.01 vs OHP
ANOVA post test Dunnett
baseline
end of treatment
3ws FU
6ws FU
9ws FU

Plantar test

At the end of treatment OHP induced hypoalgesia in all groups compared to control mice.
At 3 weeks FU the OHP and placebo groups, but not the PGN-503 mice, developed hyperalgesia at Plantar test, which recovered by 9 weeks FU.

Materials and Methods

General toxicity: the general condition of animal was evaluated daily and the body weight was measured twice a week (data not shown).
Neurophysiology: sensory NCV were evaluated stimulating the digital nerve. NCV were calculated from the latency of the stimulus artifact to the onset of the first peak of the elicited action potential and the distance between the recording and the stimulating points. AMP was calculated peak-to-peak.
Dynamic Test: after the acclimatization period, a mechanical stimulus was applied to the plantar surface of the hind paw, which exerted a progressively increasing punctuate pressure, reaching up to 15 g within 15 seconds. The pressure evoking a clear voluntary hind-paw withdrawal response was recorded automatically and taken as representing the mechanical nociceptive threshold index.
Plantar Test: after the acclimation period an infra-red light source (IR40) was positioned under the center of the mice's rear paw. On paw withdrawal, a photo cell automatically shut off the heat source and recorded the time to withdrawal (withdrawal latency).
Statistical analysis: ANOVA post test Dunnet.

Conclusions

PGN-503 significantly accelerates recovery of Oxaliplatin-induced painful peripheral neuropathy by:

- improving nerve conduction
- ameliorating mechanical allodynia and thermal hyperalgesia.

Sin
XLVII CONGRESSO NAZIONALE
22-25 OTTOBRE 2016 – VENEZIA