Abstract No. 416

Administration of IFN beta 1a in the evening ameliorates flu-like symptoms in the early phase of treatment in multiple sclerosis: data from RELIEF study

F. Patti [1], G. Zimatore [2], V. Brescia Morra [3], U. Aguglia [4], R. Bruno Bossio [5], S. Stecchi [6], L. Capone [7], R. Marziolo [8], P. Valentino [9], S. Messina [1], F. Torelli [10], M. Zappia [1]

[1]Neurology Department 1 – University Hospital "Rodolico" - Catania; [2]Neurology Department - Hospital "Dimiccoli" - Barletta; [3]Neurology Department - University Hospital "Federico II" – Naples; [4]Neurology Department - Hospital "Bianchi Melacrino Morelli" - Reggio Calabria; [5]Neurology Department – Multiple Sclerosis Center ASP – Cosenza; [6]Multiple Sclerosis Center - Villa Mazzacorati - Bologna; [7]Neurology Department – Hospital "degli Infermi" - Biella; [8]Neurology Department – Emergency Hospital "Cannizzaro" - Catania; [9]Neurology Department – University Hospital "Località Germaneto" – Catanzaro; [10]Medical Affairs – Merck Serono SpA – Rome

INTRODUCTION

- Flu-like symptoms (FLS) are among the most common adverse effects in the use of interferon (IFN) beta in multiple sclerosis (MS)
- Although it is recommended that IFN-beta injections should be administered in the evening, previous studies had reported that morning injections could reduce severity of FLS due to a decrease in interleukin-6 secretion [1], [2]

OBJECTIVE

- The primary objective was to evaluate whether administration of subcutaneous IFN beta 1a (Rebif) in the morning or in the evening may affect the severity of FLS and patient-perceived symptoms in subjects with relapsing MS over a period of 12 weeks
- The primary endpoint was to assess the severity of FLS, as measured by items 13-16 of the Multiple Sclerosis Treatment Concern Questionnaire (MSTCQ), in subjects injecting Rebif 44 mcg thrice weekly in the morning compared to the evening administration
- The main secondary objectives were aimed to assess the longitudinal changes in injection site reactions and global side-effects, changes in anxiety symptoms, fatigue symptoms, sleep disorders and quality of life in the morning group compared to the evening and to assess adherence to treatment

Figure 1. Patients disposition

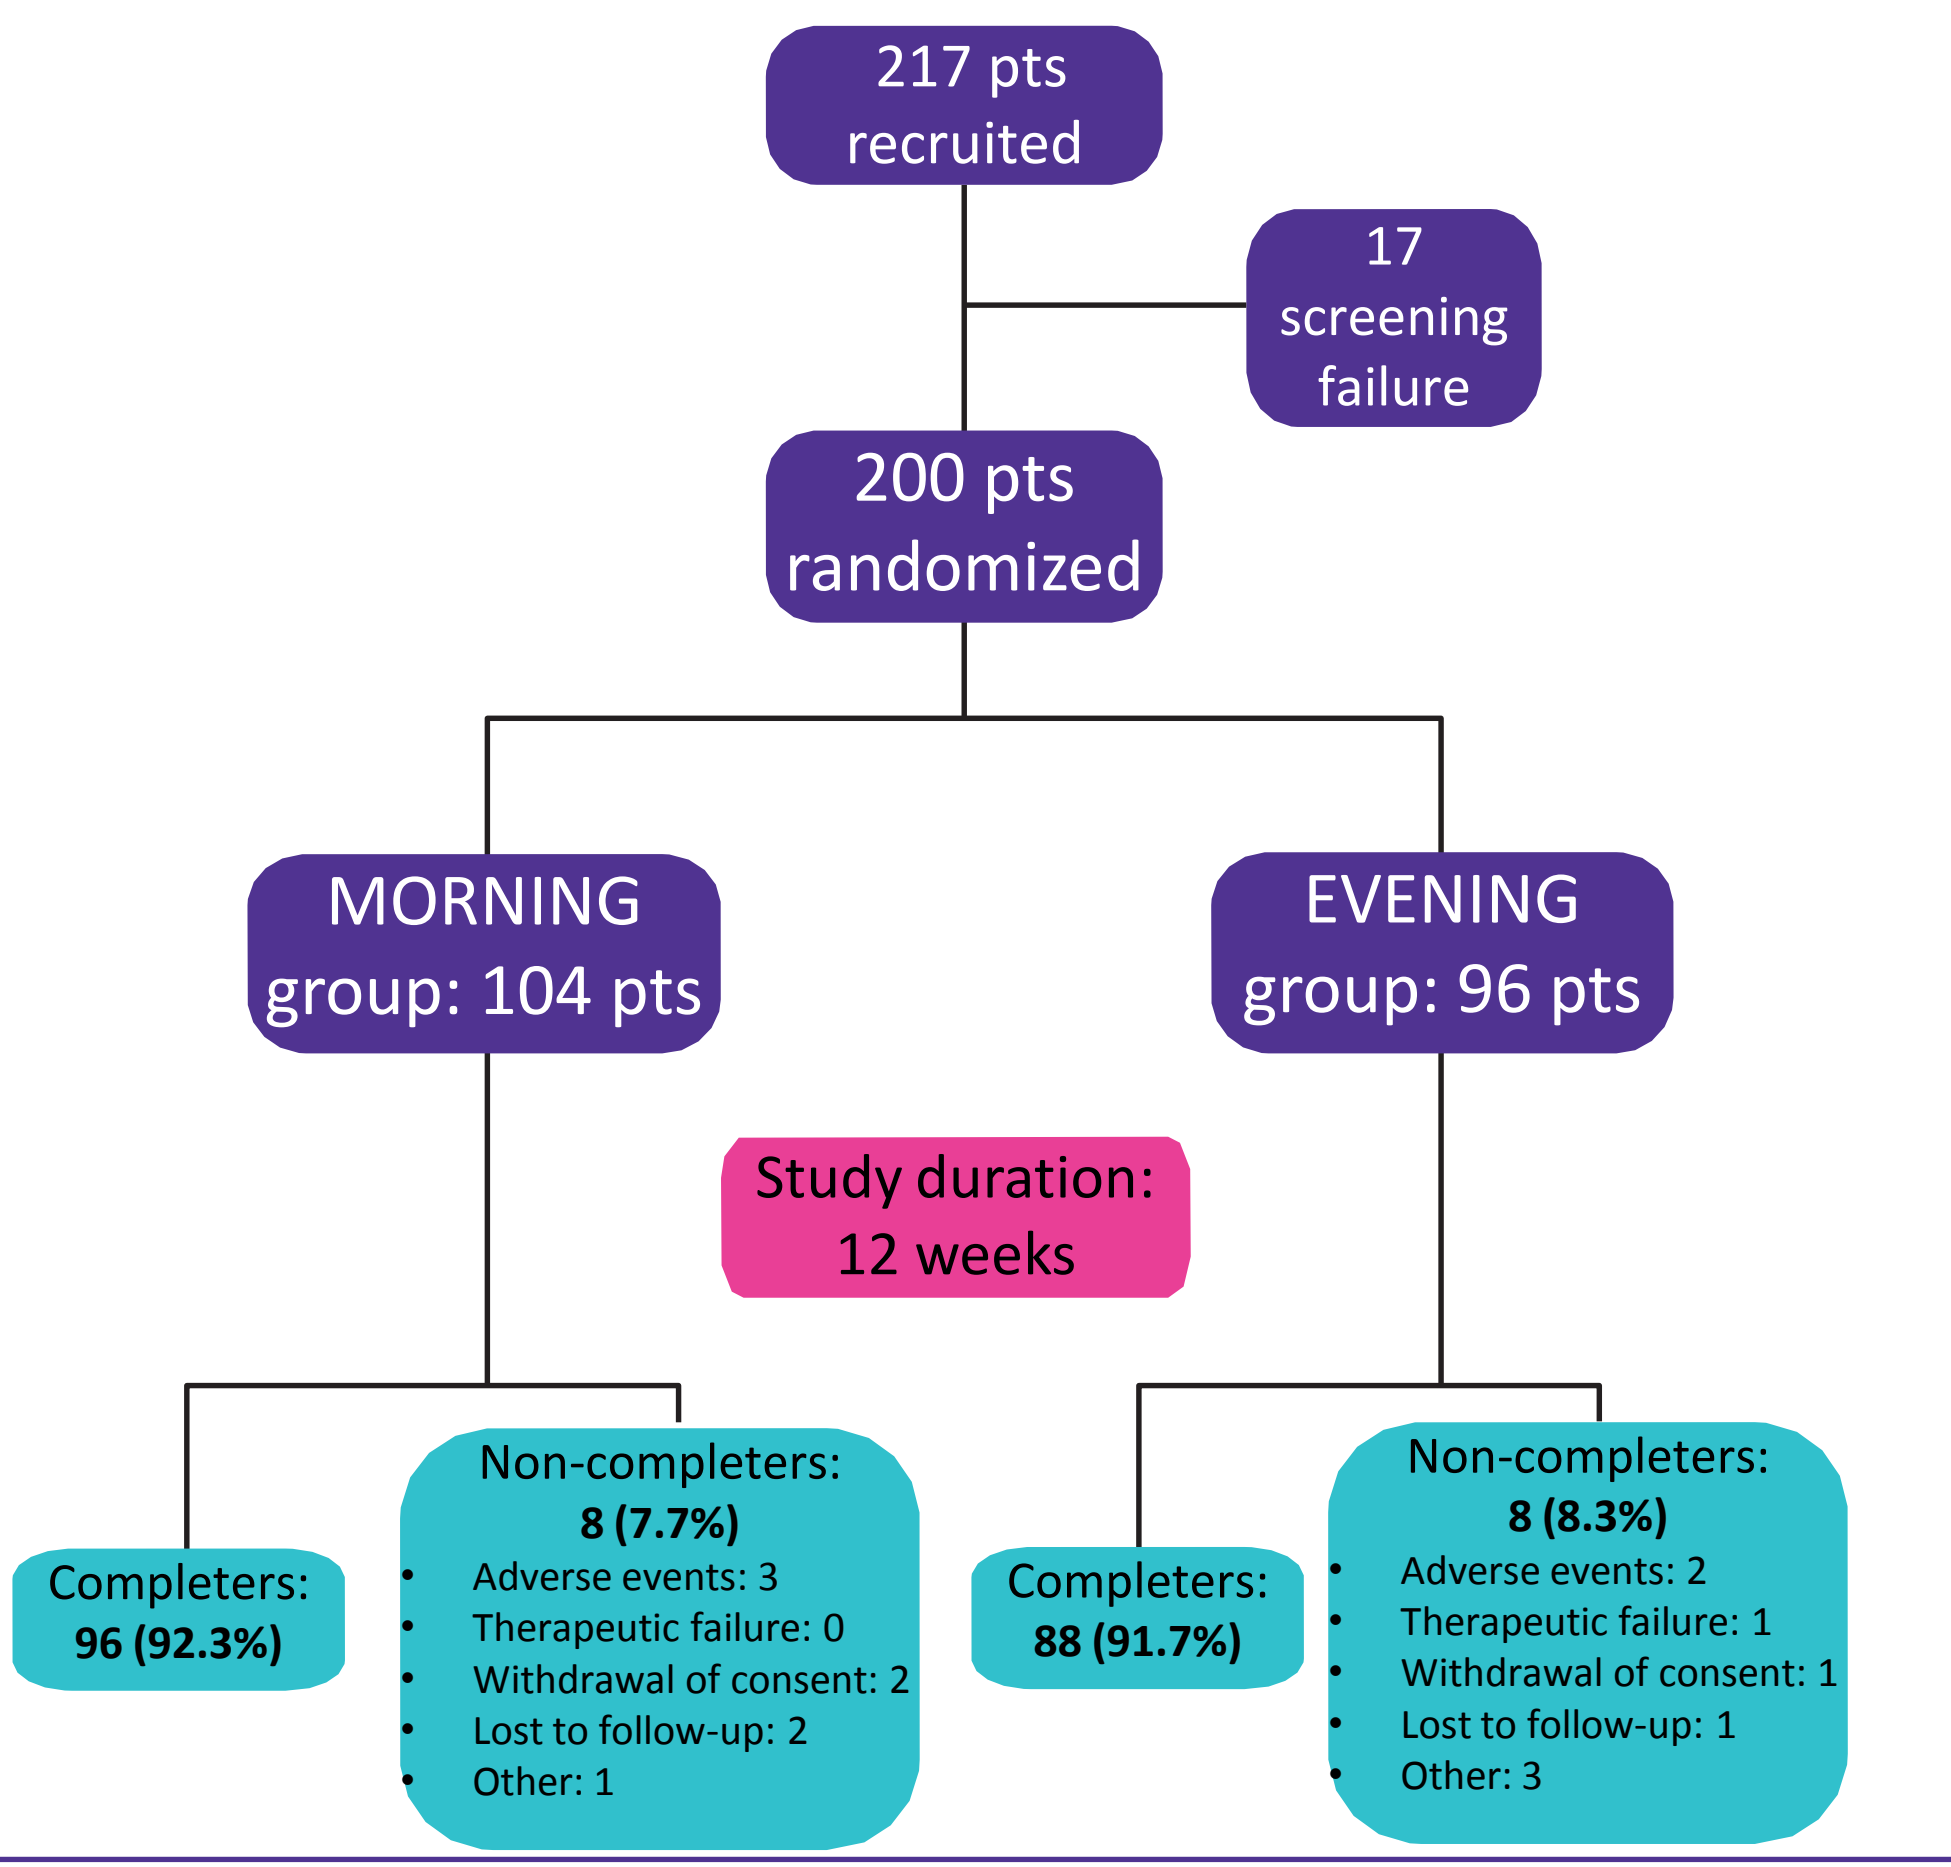
217 pts recruited
17 screening failure
200 pts randomized
MORNING group: 104 pts
EVENING group: 96 pts
Study duration: 12 weeks
Completers: 96 (92.3%)
Non-completers: 8 (7.7%)
Adverse events: 3
Therapeutic failure: 0
Withdrawal of consent: 2
Lost to follow-up: 2
Other: 1
Completers: 88 (91.7%)
Non-completers: 8 (8.3%)
Adverse events: 2
Therapeutic failure: 1
Withdrawal of consent: 1
Lost to follow-up: 1
Other: 3

METHODS

Study Design

RELIEF was a multicenter, open-label, prospective, randomized study

Figure 2. Study flow-chart

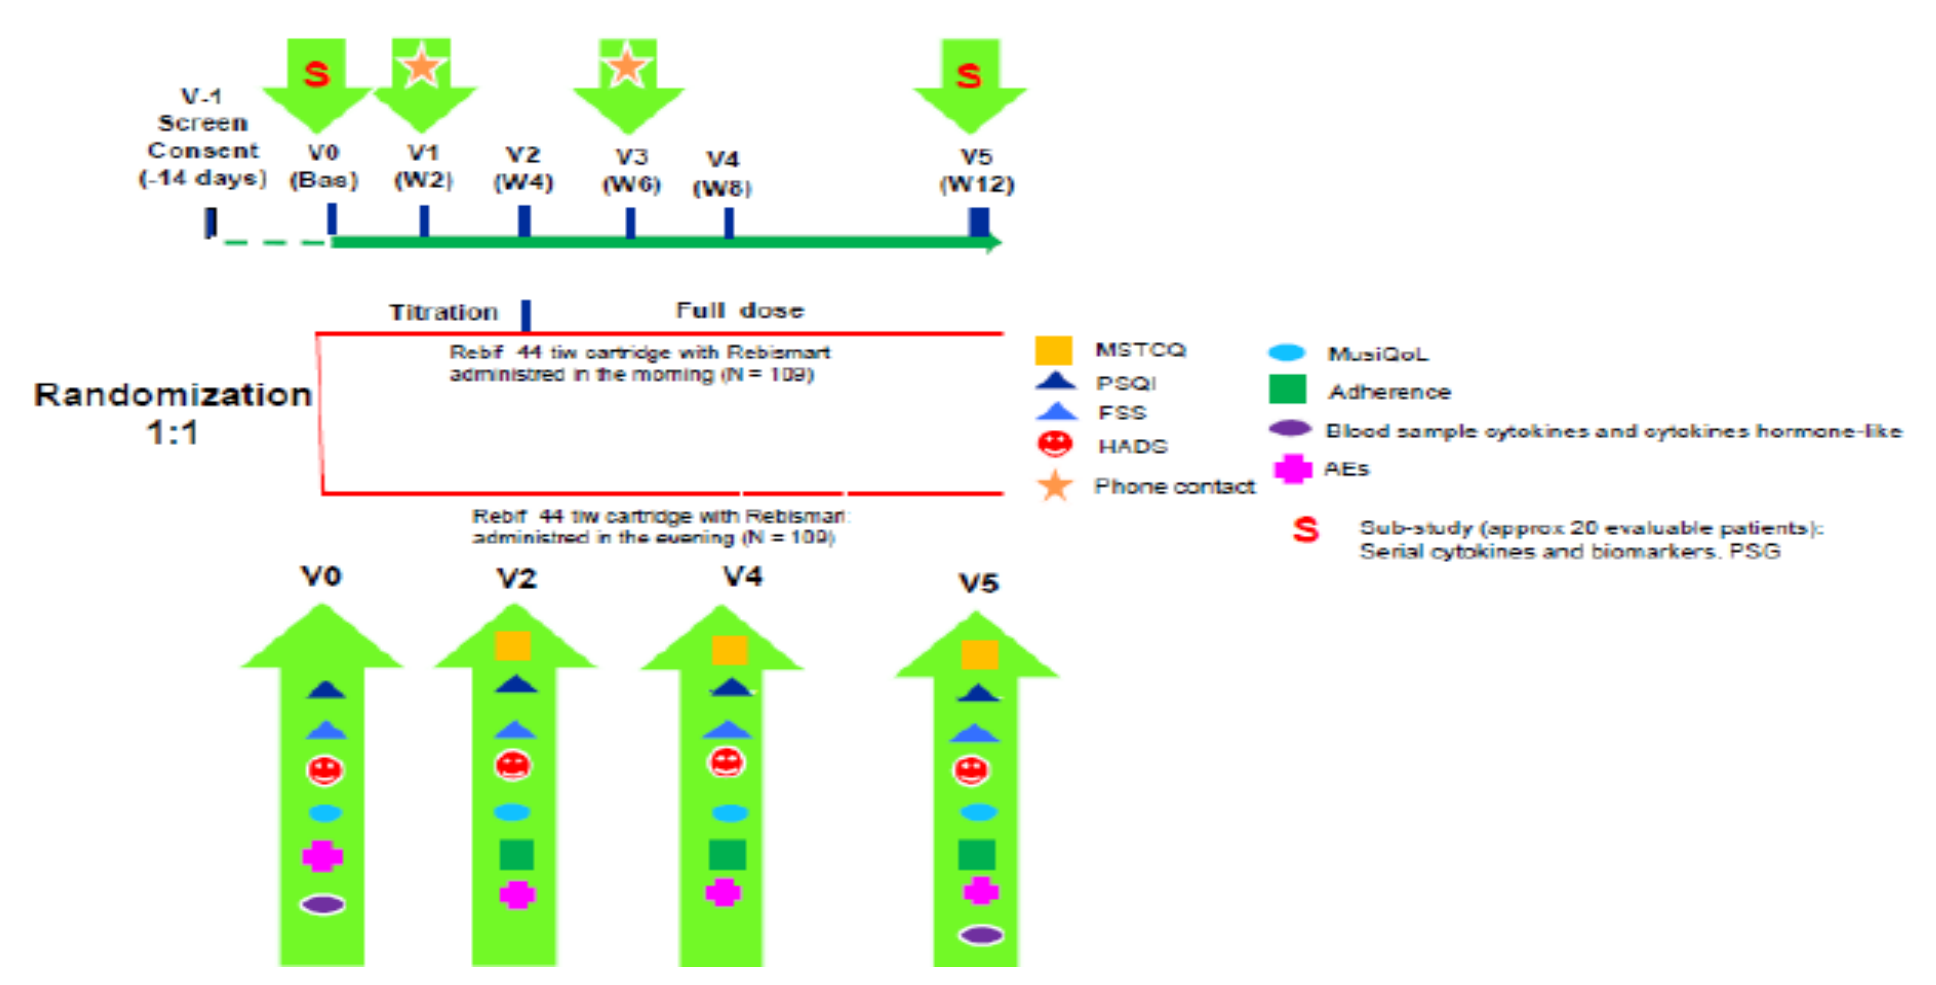
V-1 Screen Consent (-14 days)
V0 (Bas)
V1 (W2)
V2 (W4)
V3 (W6)
V4 (W8)
V5 (W12)
Titration
Full dose
Randomization 1:1
MSTCQ
PSQI
FSS
HADS
Phone contact
MusiQoL
Adherence
Blood sample cytokines and cytokines hormone-like
AEs
V0
V2
V4
V5

Evaluations (Figure 2)

- MSTCQ
- Hospital Anxiety and Depression Scale (HADS)
- Fatigue Severity Scale (FSS)
- Pittsburgh Sleep Quality Index (PSQI)
- Multiple Sclerosis International Quality of Life
- Adherence to treatment using RebiSmart®
- Circulating levels of cytokines

RESULTS

Study population

217 patients were included in the study at 29 Italian sites and 200 were randomized. Of these, 104 patients (76 females and 28 males, mean age 37.3 ± 10.1 years, mean baseline EDSS 1.89) were randomized to administer IFN beta 1a in the morning and 96 patients (62 females and 34 males, mean age 34.9 ± 10.8 years, mean baseline EDSS 1.87) in the evening **(Figure 1)**

Main Analysis results (Figure 3)

- **4 weeks** after starting therapy, patients in the morning group reported a significantly higher score in items 13-16 of MSTCQ when compared to the evening group (difference between adjusted means: 1.35, CI 0.35-2.35, p=0.008)
- The difference was still significant after **8 weeks** (difference between adjusted means: 1.34, CI 0.35-2.32, p=0.008)
- The difference was no longer significant at the end of the observation period of **12 weeks** of therapy (difference between adjusted means: 0.47, CI -0.52-1.46, p=0.35)

Figure 3. MSTCQ: FLS scores evening *vs* morning

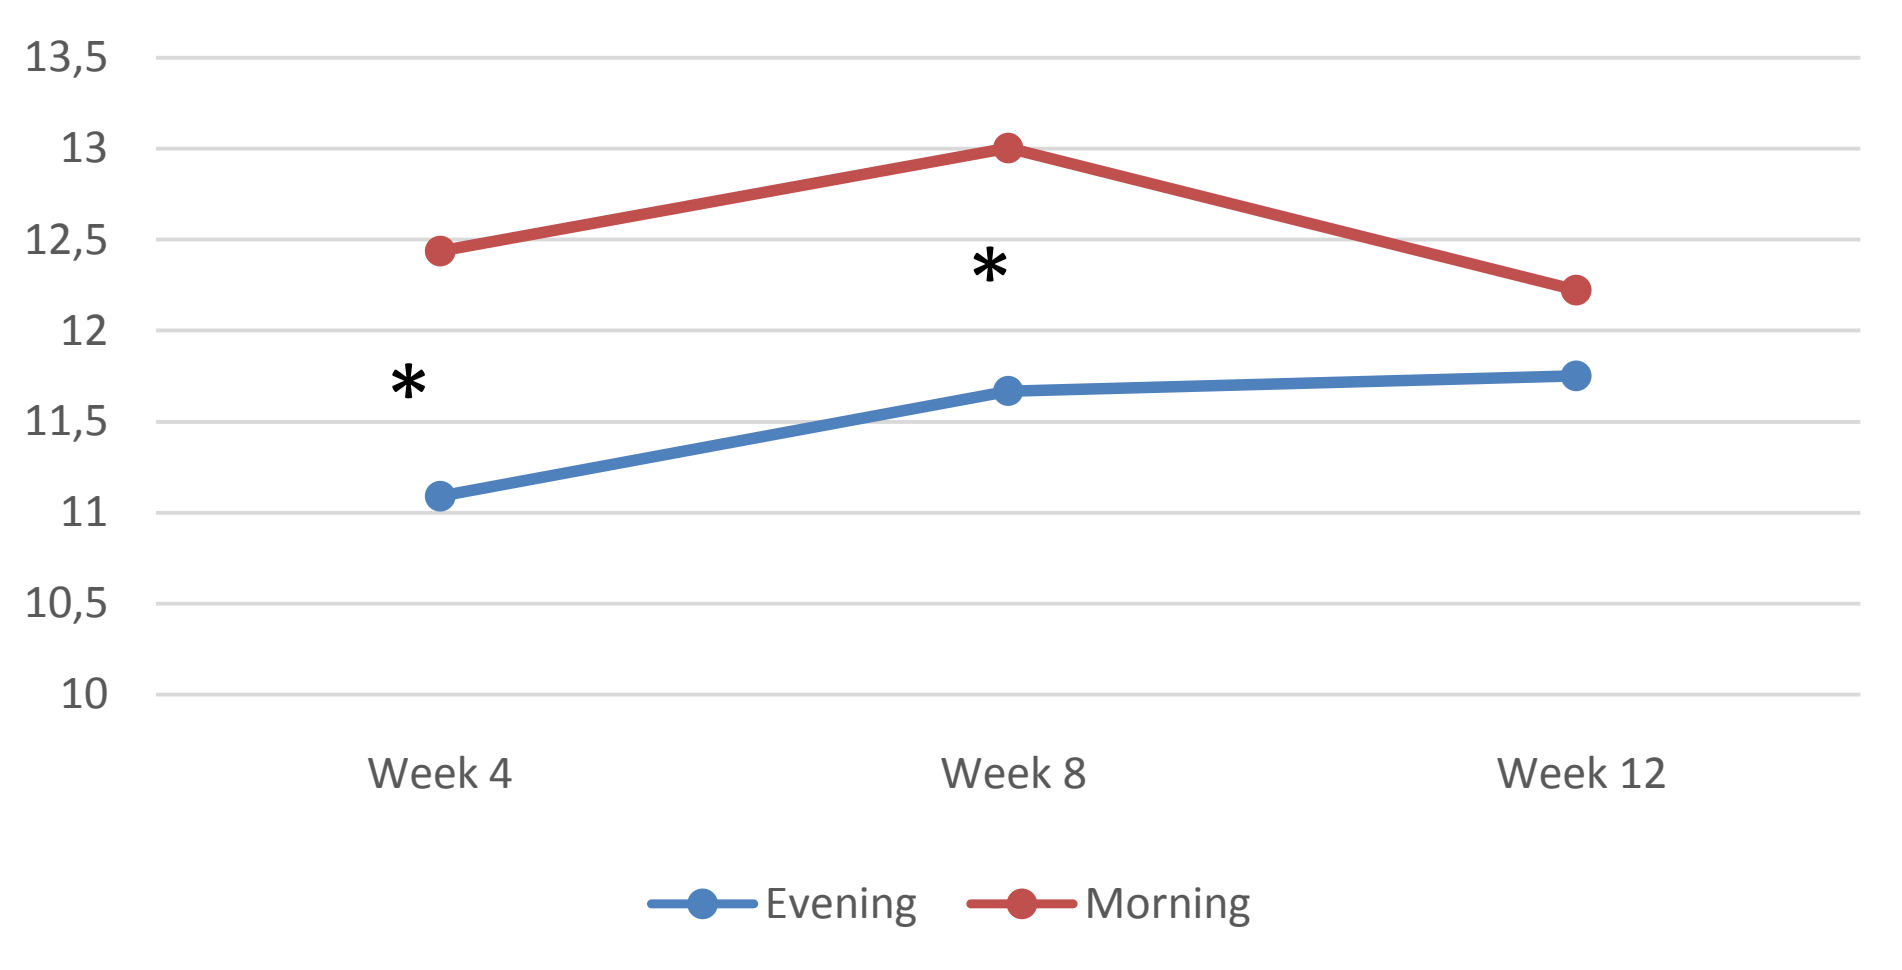
13,5
13
12,5
12
11,5
11
10,5
10
Week 4
Week 8
Week 12
*
*
Evening
Morning

* p value for adjusted mean difference Morning - Evening < 0.01.
Analysis is based on a linear mixed model for repeated measures with FLS score at each visit as dependent variable, while treatment group, visit (Week 4, Week 8, Week 12), treatment by visit interaction as fixed factors of the model and patient as random effect

Secondary Analysis

Adherence to study medication

Mean (±SD) cumulative adherence recorded by RebiSmart® at week 12 was 98.6 ± 9.8 % in the morning group and 98.1 ± 10.0 % in the evening group.
(Adherence = 100 X number of injections administered/number of expected injections)

Safety results

No new safety or tolerability concerns were identified following treatment with Rebif 44 mcg tiw in the morning or evening group for 12 weeks

CONCLUSIONS

- Based on data from RELIEF study, evening injections led to an improvement of IFN beta-related flu-like symptoms in the very initial phase of treatment; with the continuation of the therapy this advantage tended to decrease
- Therefore to start the injection of IFN beta in the evening could lead to a better management of FLS in patients with MS, while in the continuation of therapy the time of administration could be decided according to patient's lifestyle and preference

REFERENCES

1. Kümpfel T, Schwan M, Pollmächer T, Yassouridis A, Uhr M, Trenkwalder C, Weber F. Time of interferon-beta 1a injection and duration of treatment affect clinical side effects and acute changes of plasma hormone and cytokine levels in multiple sclerosis patients. Mult Scler (2007);13(9):1138-45
2. Nadjar Y, Coutelas E, Prouteau P, Panzer F, Paquet D, Saint-Val C, Créange A. Injection of interferon-beta in the morning decreases flu-like syndrome in many patients with multiple sclerosis. Clin Neurol Neurosurg (2011);113(4):316-322

ACKNOWLEDGMENTS

This trial was sponsored by Merck Serono S.p.A. (Italy), an affiliate of Merck KGaA, Darmstadt, Germany. The authors would like to thank the patients, investigators, co-investigators, and the study teams at each of the participating centers.
Correspondence: *patti@unict.it*

DISCLOSURES

Prof. Patti served on the scientific advisory board for Teva, Biogen-Idec, Bayer-Schering, Novartis, and has received honoraria as a speaker for Teva, Biogen, Merck-Serono, Bayer-Schering, Genzyme/Sanofi, and Novartis.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at Italian Neurological Society (SIN) Congress; October 14-17; Naples, Italy